Phase 3 Clinical Studies of ZX008 (Fenfluramine Hydrochloride) Oral Solution as an Adjunctive Therapy for Seizures in Children and Young Adults with Dravet Syndrome

Joseph Sullivan,[1] Lieven Lagae,[2] Kevin Romanko,[3] Rosemary Kerwin,[3] Anita Maiers,[3] Bradley Galer,[3] Gail Farfel[3]

[1]University of California, San Francisco, San Francisco, CA, USA; [2]University of Leuven, Leuven, Belgium; [3]Zogenix, Inc., San Diego, CA, USA

INTRODUCTION

- A retrospective chart review and small, prospective, observational study have suggested that fenfluramine has antiepileptic activity in patients with Dravet syndrome (DS)[1-3]
- Based on data from these open-label investigations, Zogenix is initiating a Phase 3 development program for ZX008 (fenfluramine HCl oral suspension) as adjunctive treatment for uncontrolled seizures in children with DS
- Here we present an outline of the Phase 3 clinical study program for ZX008 as adjunctive treatment of seizures in children with DS
 - Two identical, double-blind, placebo-controlled, fixed-dose, acute-care studies
 - Study 1501 in United States and Canada – initiation in late 2015
 - Study 1502 in Europe, Australia, and other countries – initiation in 2016
 - Patients in both studies will be eligible to enroll in an open-label, long-term extension study (**Figure 1**)

Figure 1. ZX008 Clinical Development Plan

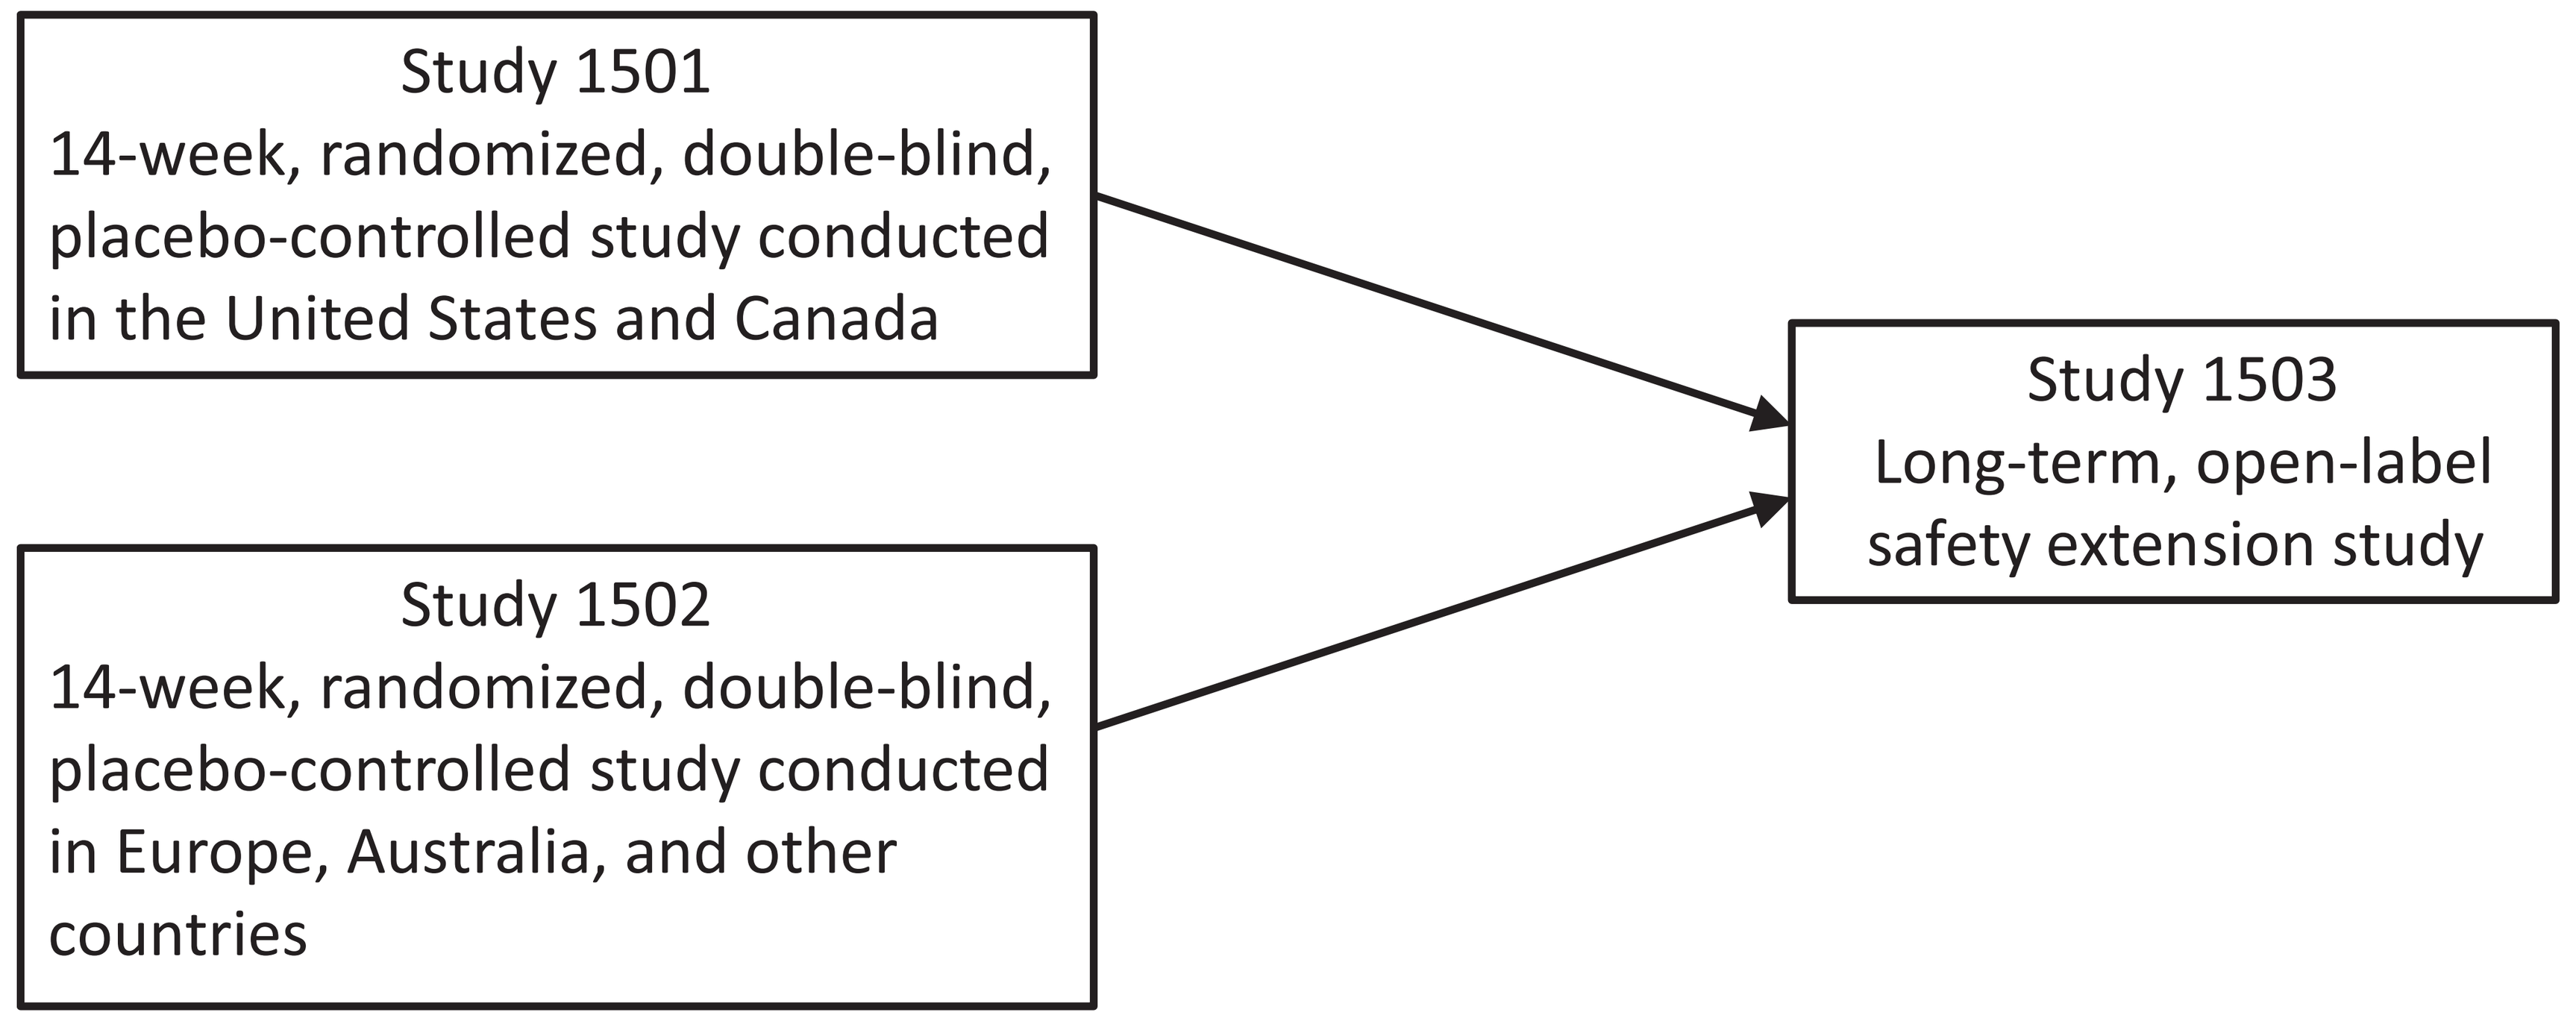
Study 1501
14-week, randomized, double-blind, placebo-controlled study conducted in the United States and Canada
Study 1502
14-week, randomized, double-blind, placebo-controlled study conducted in Europe, Australia, and other countries
Study 1503
Long-term, open-label safety extension study

STUDIES 1501 AND 1502: PROPOSED PROTOCOL

Subjects

Key Inclusion Criteria

- Male or female subjects age ≥2 to ≤18 years with a clinical diagnosis of DS with >6 convulsive seizures in the baseline period
- Subject must meet **all 5** of the following criteria:
 - Onset of seizures in first year of life
 - History of prolonged convulsive seizures
 - Initial development was normal
 - Normal brain MRI without cortical malformation
 - Lack of an alternative diagnosis
- Subject must meet **1** of the following criteria:
 - Emergence of a second seizure type, after the first seizure type
 - Fever or exposure to warm temperatures or increased core body temperature induces seizures
 - Has photosensitive or pattern-sensitive seizures
 - Genetic test results are consistent with a diagnosis of DS
- All medication or interventions for epilepsy are stable and are expected to remain stable during study

Key Exclusion Criteria

- Pregnant or lactating females
- History of cardiovascular disease, pulmonary hypertension, anorexia nervosa, depression, or glaucoma
- Treatment with concomitant serotonergic medications
- Treatment with cytochrome P450 2D6/3A4/2B6 inhibitors or substrates
- Treatment with stiripentol in past 21 days before screening, or participation in an experimental protocol in past 30 days
- Current treatment with carbamazepine, oxcarbazepine, phenobarbital, phenytoin, or any cannabinoid product, or treatment with any of these in the previous 30 days as maintenance therapy

STUDY DESIGN FOR ACUTE PHASE 3 EFFICACY/SAFETY TRIALS

- Multicenter, randomized, double-blind, parallel-group, placebo-controlled
- Pediatric and young adult subjects with DS (N=105 in each study)
 - Stratified by age: <6 years old, ≥6 to 18 years old
- **Figure 2** presents the Acute Phase 3 Trial Study Design
- Experimental treatments
 - ZX008 oral solution 0.2 mg/kg/day
 - ZX008 oral solution 0.8 mg/kg/day (max 30 mg/day)
 - Placebo oral solution

Figure 2. Schematic Representation of the 2 Phase 3 Clinical Studies of ZX008 for the Treatment of Uncontrolled Seizures in Dravet Syndrome Patients

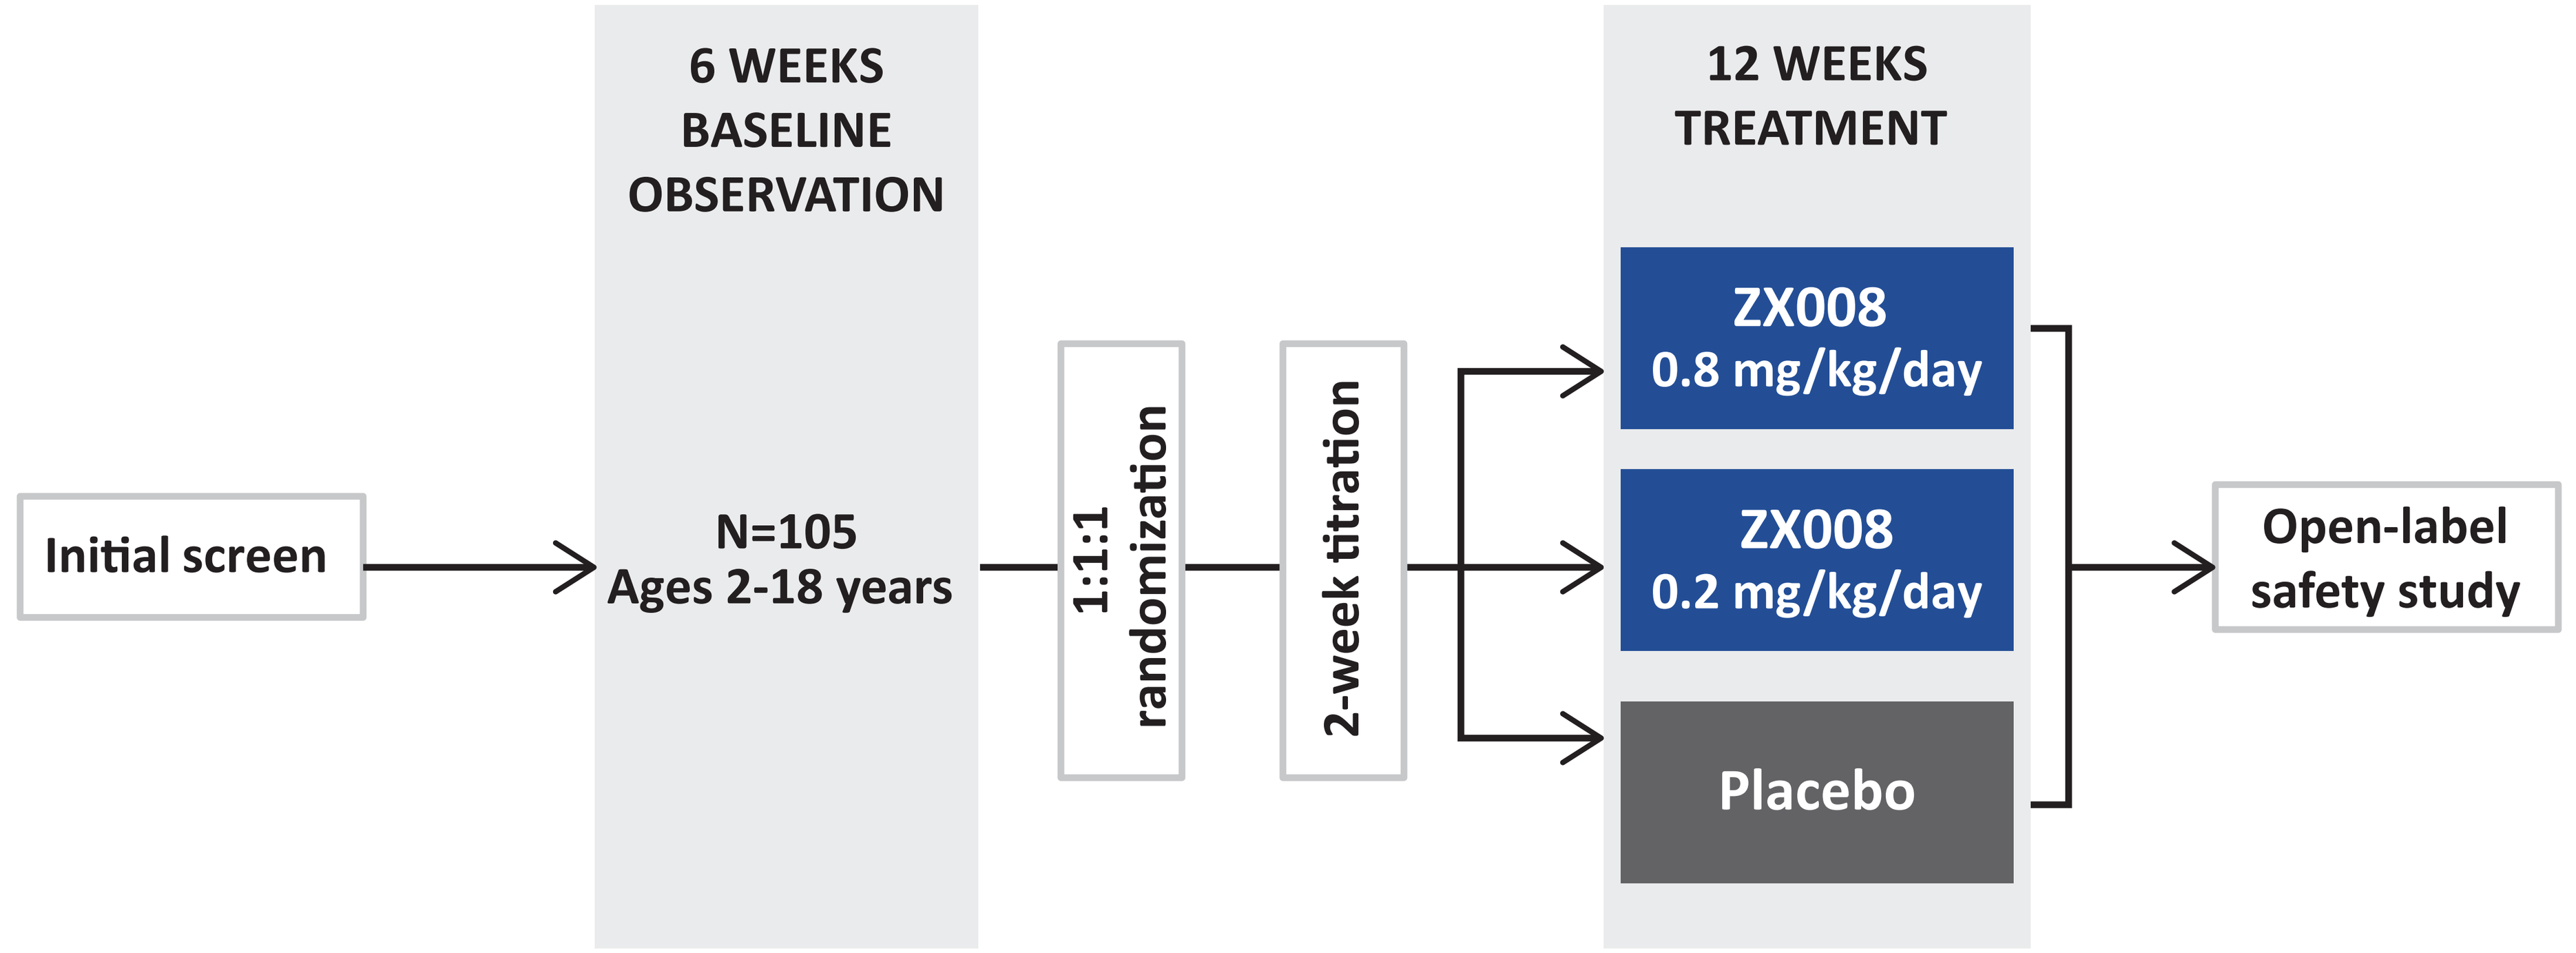
6 WEEKS BASELINE OBSERVATION
12 WEEKS TREATMENT
Initial screen
N=105
Ages 2-18 years
1:1:1 randomization
2-week titration
ZX008
0.8 mg/kg/day
ZX008
0.2 mg/kg/day
Placebo
Open-label safety study

Key Efficacy Assessments

- Primary analysis will compare ZX008 0.8 mg/kg/day vs placebo
 - Incidence of convulsive seizures during double-blind treatment period
- Planned secondary analyses include:
 - Comparison of ZX008 0.2 mg/kg/day with placebo and ZX008 0.8 mg/kg/day on incidence of convulsive seizures during double-blind treatment period
 - Longest convulsive seizure-free interval
 - Proportion of patients who achieve a ≥40% or ≥50% reduction in convulsive seizure frequency
 - Clinical Global Impression – improvement as assessed by parent/caregiver and by investigator

Key Safety Assessments

- Adverse events
- Laboratory safety parameters (hematology, chemistry, urinalysis)
- Vital signs
- Cardiac examination
 - Doppler echocardiogram
 - 12-lead electrocardiogram

SUMMARY

- The proposed 2 randomized, double-blind studies of ZX008 oral solution at doses of 0.8 and 0.2 mg/kg/day compared with placebo, plus long-term, open-label safety extension are designed to provide adequate efficacy and safety information for registration for the indication of adjunctive treatment of uncontrolled seizures in pediatric patients with DS

REFERENCES

1. Ceulemans B, et al. Successful use of fenfluramine as an add-on treatment for Dravet syndrome. *Epilepsia*. 2012;53:1131-9.
2. Ceulemans B, et al. Low-dose fenfluramine significantly reduces seizure frequency in Dravet syndrome: a prospective study of a new cohort of Belgian patients. Presented at the 69th Annual Meeting of the American Epilepsy Society; Philadelphia, PA, 2015: Poster 2.262.
3. Schoonjans A-S, et al. Cardiovascular side effects of low-dose fenfluramine treatment in Dravet syndrome: a prospective echocardiographic pilot study. Presented at the 69th Annual Meeting of the American Epilepsy Society; Philadelphia, PA, 2015: Poster 2.266.

ACKNOWLEDGMENTS

These clinical studies are being sponsored by Zogenix, Inc. Medical writing assistance, medical editing, and poster production was provided by PharmaWrite, LLC (Princeton, NJ) and was funded by Zogenix, Inc.

DISCLOSURE

The studies presented here will be conducted by Zogenix, Inc. JS and LL have received honoraria as advisors and are investigators and LL has also received honoraria as a consultant from Brabant Pharma and Zogenix; RK is a contract employee and KR, AM, BG, and GF are full-time employees of Zogenix, Inc.

Presented as part of a Scientific Exhibit during the 69th Annual Meeting of the American Epilepsy Society, December 4-8, 2015, Philadelphia