2.224

Fenfluramine in Dravet Syndrome: Re-Analysis of Study 1 Using Time-to-Event to Measure Treatment Effect

ZOGENIX

Dennis J. Dlugos[1]; Jacqueline A. French[2]; Michael Lock[3]; Gail M. Farfel[3]; Douglas Haney[3]; Glenn Morrison[3]

[1]Children's Hospital of Philadelphia, Philadelphia, PA, USA; [2]NYU Comprehensive Epilepsy Center, New York, NY, USA; [3]Zogenix, Inc., Emeryville, CA, USA

Introduction

- Randomized, placebo-controlled clinical studies are considered the "gold standard" of contemporary studies of antiepileptic drugs (AEDs)
 - Typically have a 4- to 8-week baseline period to establish seizure frequency
 - Titration period plus 12-week fixed-dose treatment period
 - Primary analysis is change from baseline in seizure frequency
- For patients with epileptic encephalopathies, these studies can be problematic
 - Long baseline period
 - Long duration of exposure to placebo
- A "time-to-event" (TTE) study design has been proposed to address these limitations
 - Minimize duration of placebo exposure
 - Reduce the overall study length
 - Remain sufficiently sensitive to detect a treatment effect
 - May allow a reduction in baseline duration
- Here we present a proof-of-concept analysis using data acquired in Study 1, a randomized, placebo-controlled clinical study that followed the "gold standard" study design to evaluate fenfluramine for the treatment of patients with Dravet syndrome (DS)

Methods

- Study 1 (NCT02682927, NCT02826863)
 - Patients with DS whose convulsive seizures were not completely controlled by their current AED regimen were enrolled
 - Patients receiving concomitant stiripentol or cannabidiol were excluded from the study
 - 6-week baseline period to evaluate eligibility and establish baseline convulsive seizure frequency
 - Eligible patients were randomized to receive placebo or fenfluramine 0.2 or 0.7 mg/kg/day added to their stable AED regimen for 14 weeks
 - Fenfluramine was administered as ZX008, an oral solution of fenfluramine HCl containing 2.2 mg/mL fenfluramine
- In the TTE analyses, baseline seizure number was determined using 2 methods:
 - Method 1: all patients use a 1-week baseline period
 - The number of seizures occurring during the first 7 days of the Study 1 baseline period was determined for each patient
 - The number of seizures during the 7-day window was defined as the event for each patient – "Nth seizure"
 - Method 2: variable length baseline period
 - Patients were categorized by their number of seizures occurring during the first week of the original baseline period, and the duration of baseline was inversely correlated with seizure frequency
 - 0-3 seizures (low frequency): 4-week baseline
 - 4-9 seizures (moderate frequency): 2-week baseline
 - 10+ seizures (high frequency): 1-week baseline
 - Number of seizures during each patient's new baseline period was defined as the event for each patient – "Nth seizure"
 - Time-to-event
 - TTE was the duration of time in the treatment phase of the study for each patient to reach their Nth seizure
 - Data were censored for patients who did not reach their "Nth seizure" by Week 14 or time of study discontinuation
 - Differences between Kaplan-Meier TTE curves for treatment and placebo were assessed using the log-rank test

Results

- 119 patients with DS were randomized to treatment
 - Placebo: n=40
 - Fenfluramine 0.2 mg/kg/day: n=39
 - Fenfluramine 0.7 mg/kg/day: n=40
- Baseline patient characteristics are shown in **Table 1**

Table 1. Demographic and Baseline Characteristics

	Fenfluramine 0.7 mg/kg/day	Fenfluramine 0.2 mg/kg/day	Placebo
n	40	39	40
Age (years)			
Mean±SD	8.8±4.4	9.0±4.5	9.2±5.1
(Min, max)	(2, 18)	(2, 17)	(2, 18)
Age group <6 years, n (%)	11 (28)	9 (23)	11 (28)
Males, n (%)	21 (52)	22 (56)	21 (52)
BMI (kg/m^2)			
Mean±SD	18.5±3.5	19.3±5.7	18.0±3.8
Concomitant AEDs, n (%)			
Valproate (all forms)	25 (62)	24 (62)	22 (55)
Clobazam	24 (60)	24 (62)	22 (55)
Topiramate	11 (28)	10 (26)	9 (22)
Levetiracetam	4 (10)	11 (28)	11 (28)
1-week baseline seizure count, n			
0-3 (low)	16	17	11
4-9 (moderate)	13	13	13
10+ (high)	11	9	16

AED, antiepileptic drug; BMI, body mass index.

- **Figures 1-4** show the TTE analyses using the 1-week baseline seizure count and the variable length baseline seizure count

Figure 1. 1-Week Baseline Period: Kaplan-Meier Curve for Time to Nth Seizure

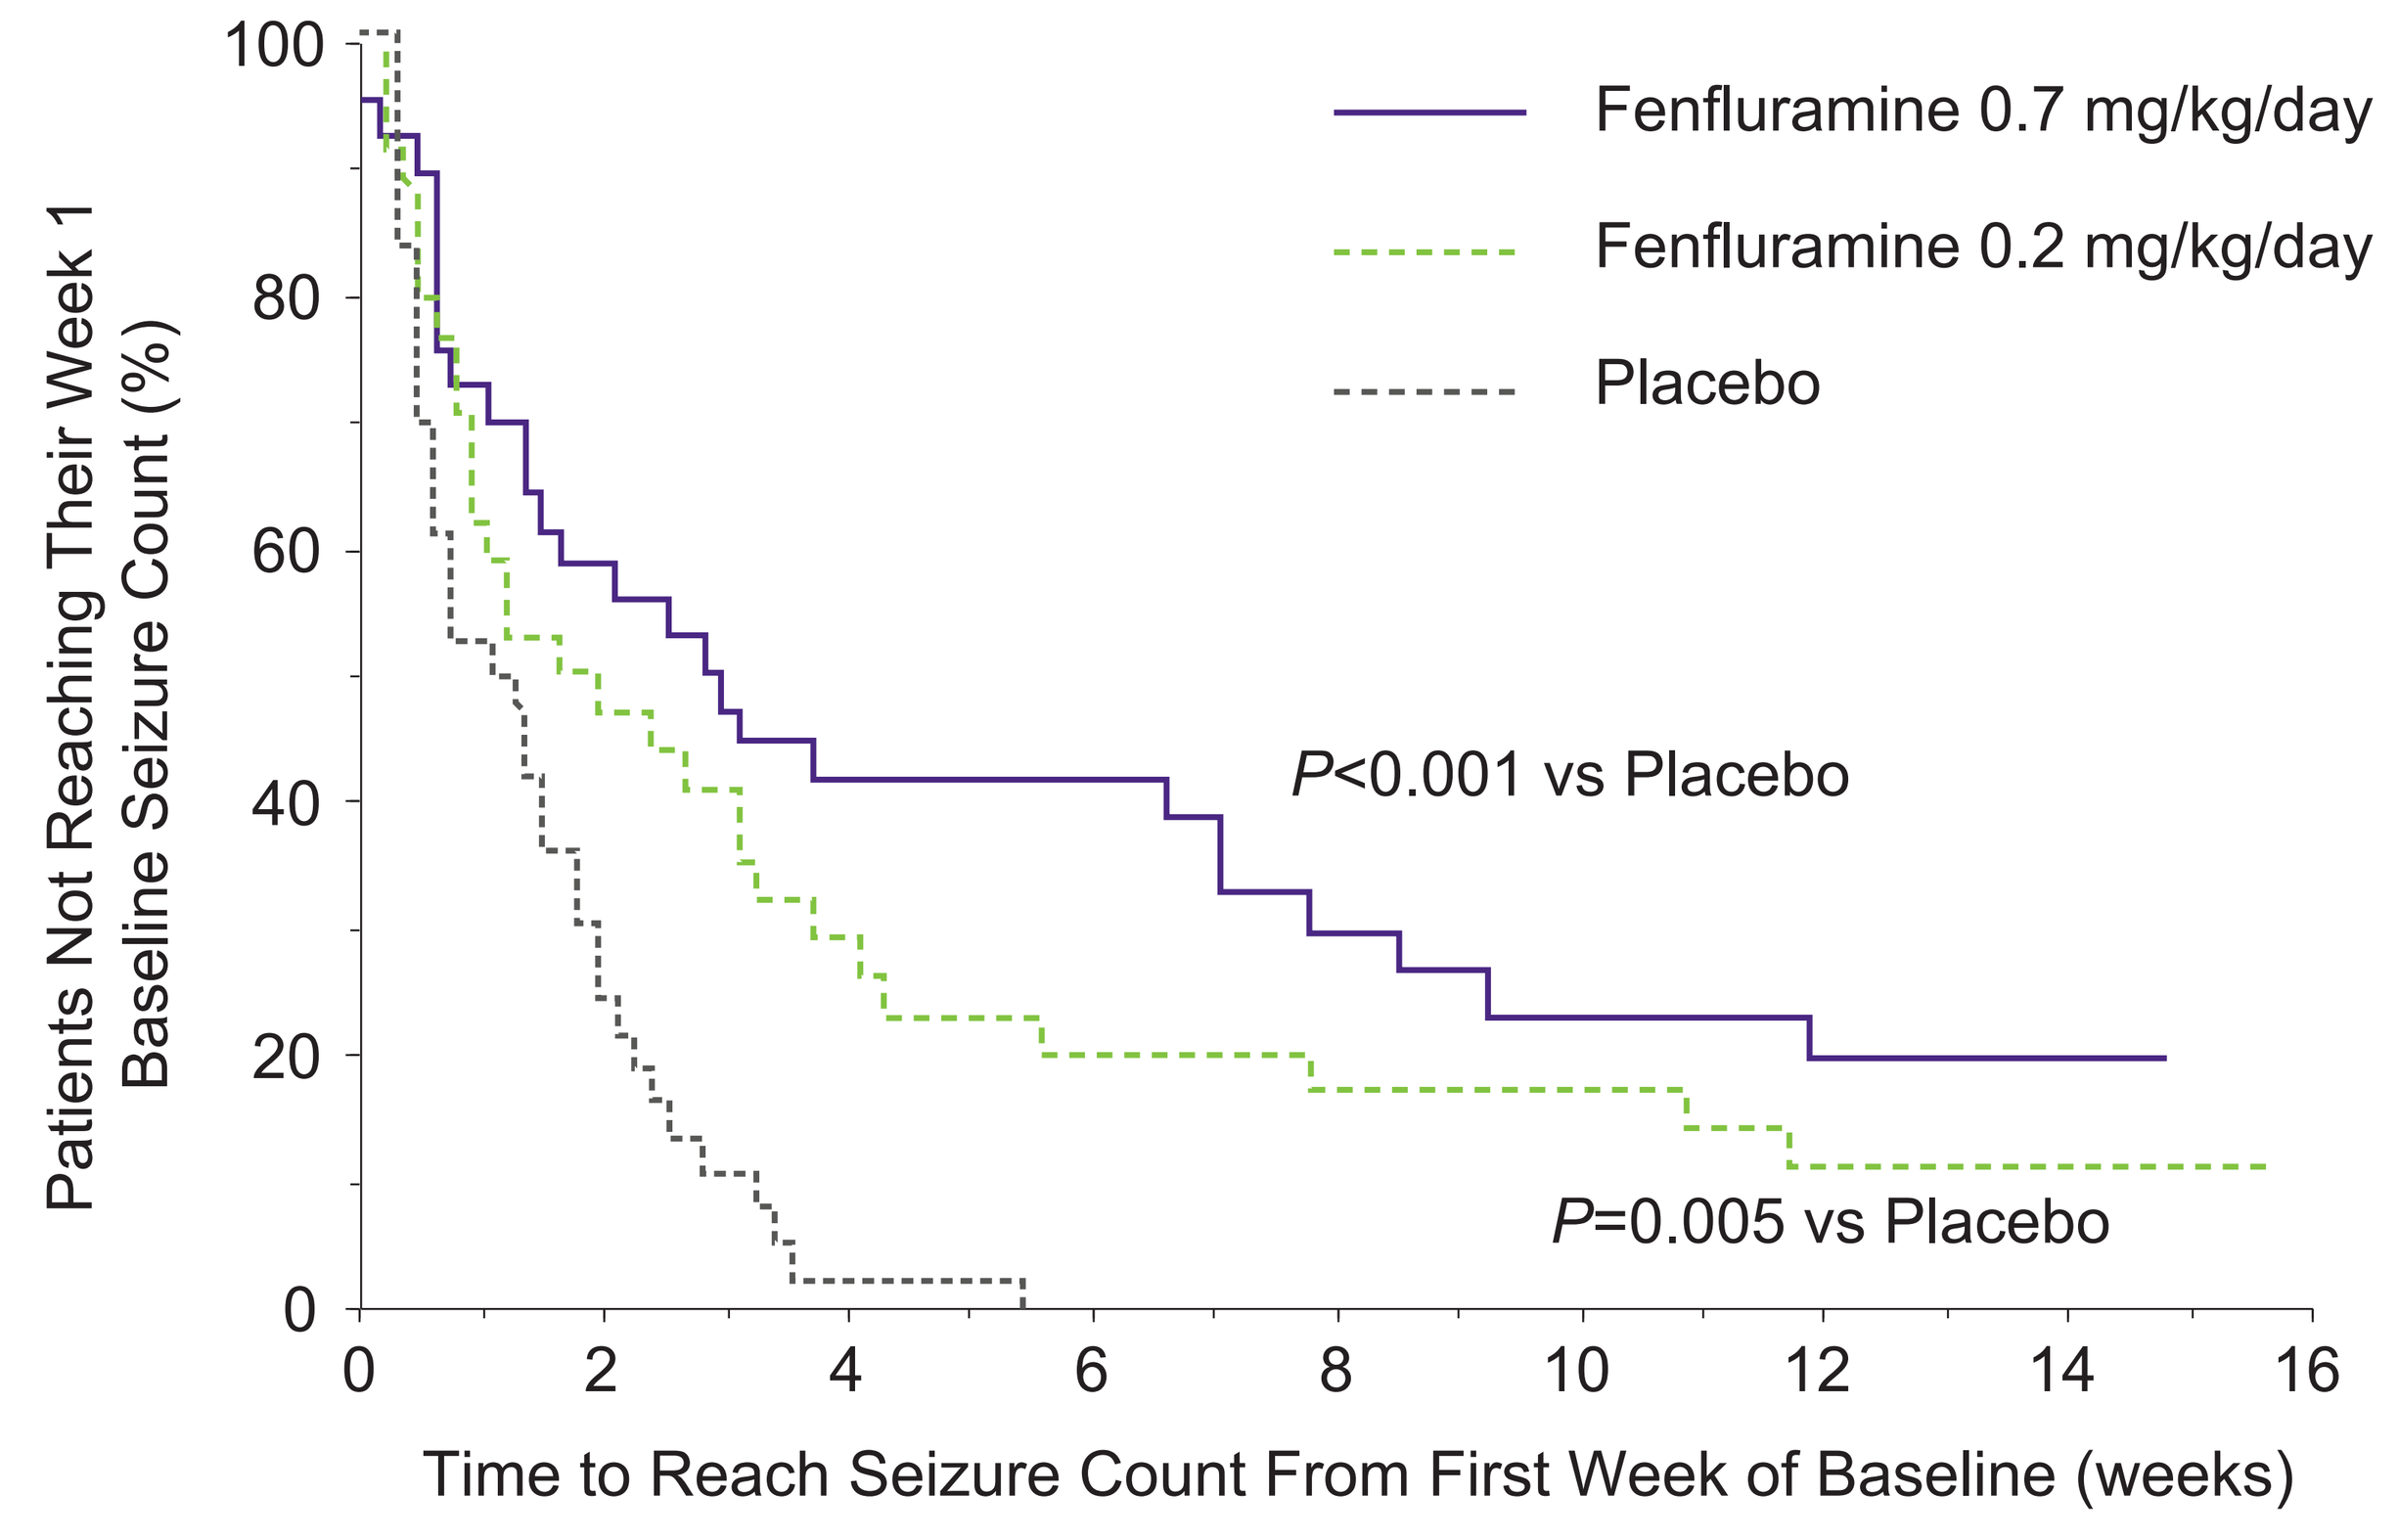

Patients Not Reaching Their Week 1 Baseline Seizure Count (%)
100
80
60
40
20
0
0
2
4
6
8
10
12
14
16
Time to Reach Seizure Count From First Week of Baseline (weeks)
Fenfluramine 0.7 mg/kg/day
Fenfluramine 0.2 mg/kg/day
Placebo
P<0.001 vs Placebo
P=0.005 vs Placebo

Figure 2. 1-Week Baseline Period: Median Time to Nth Seizure

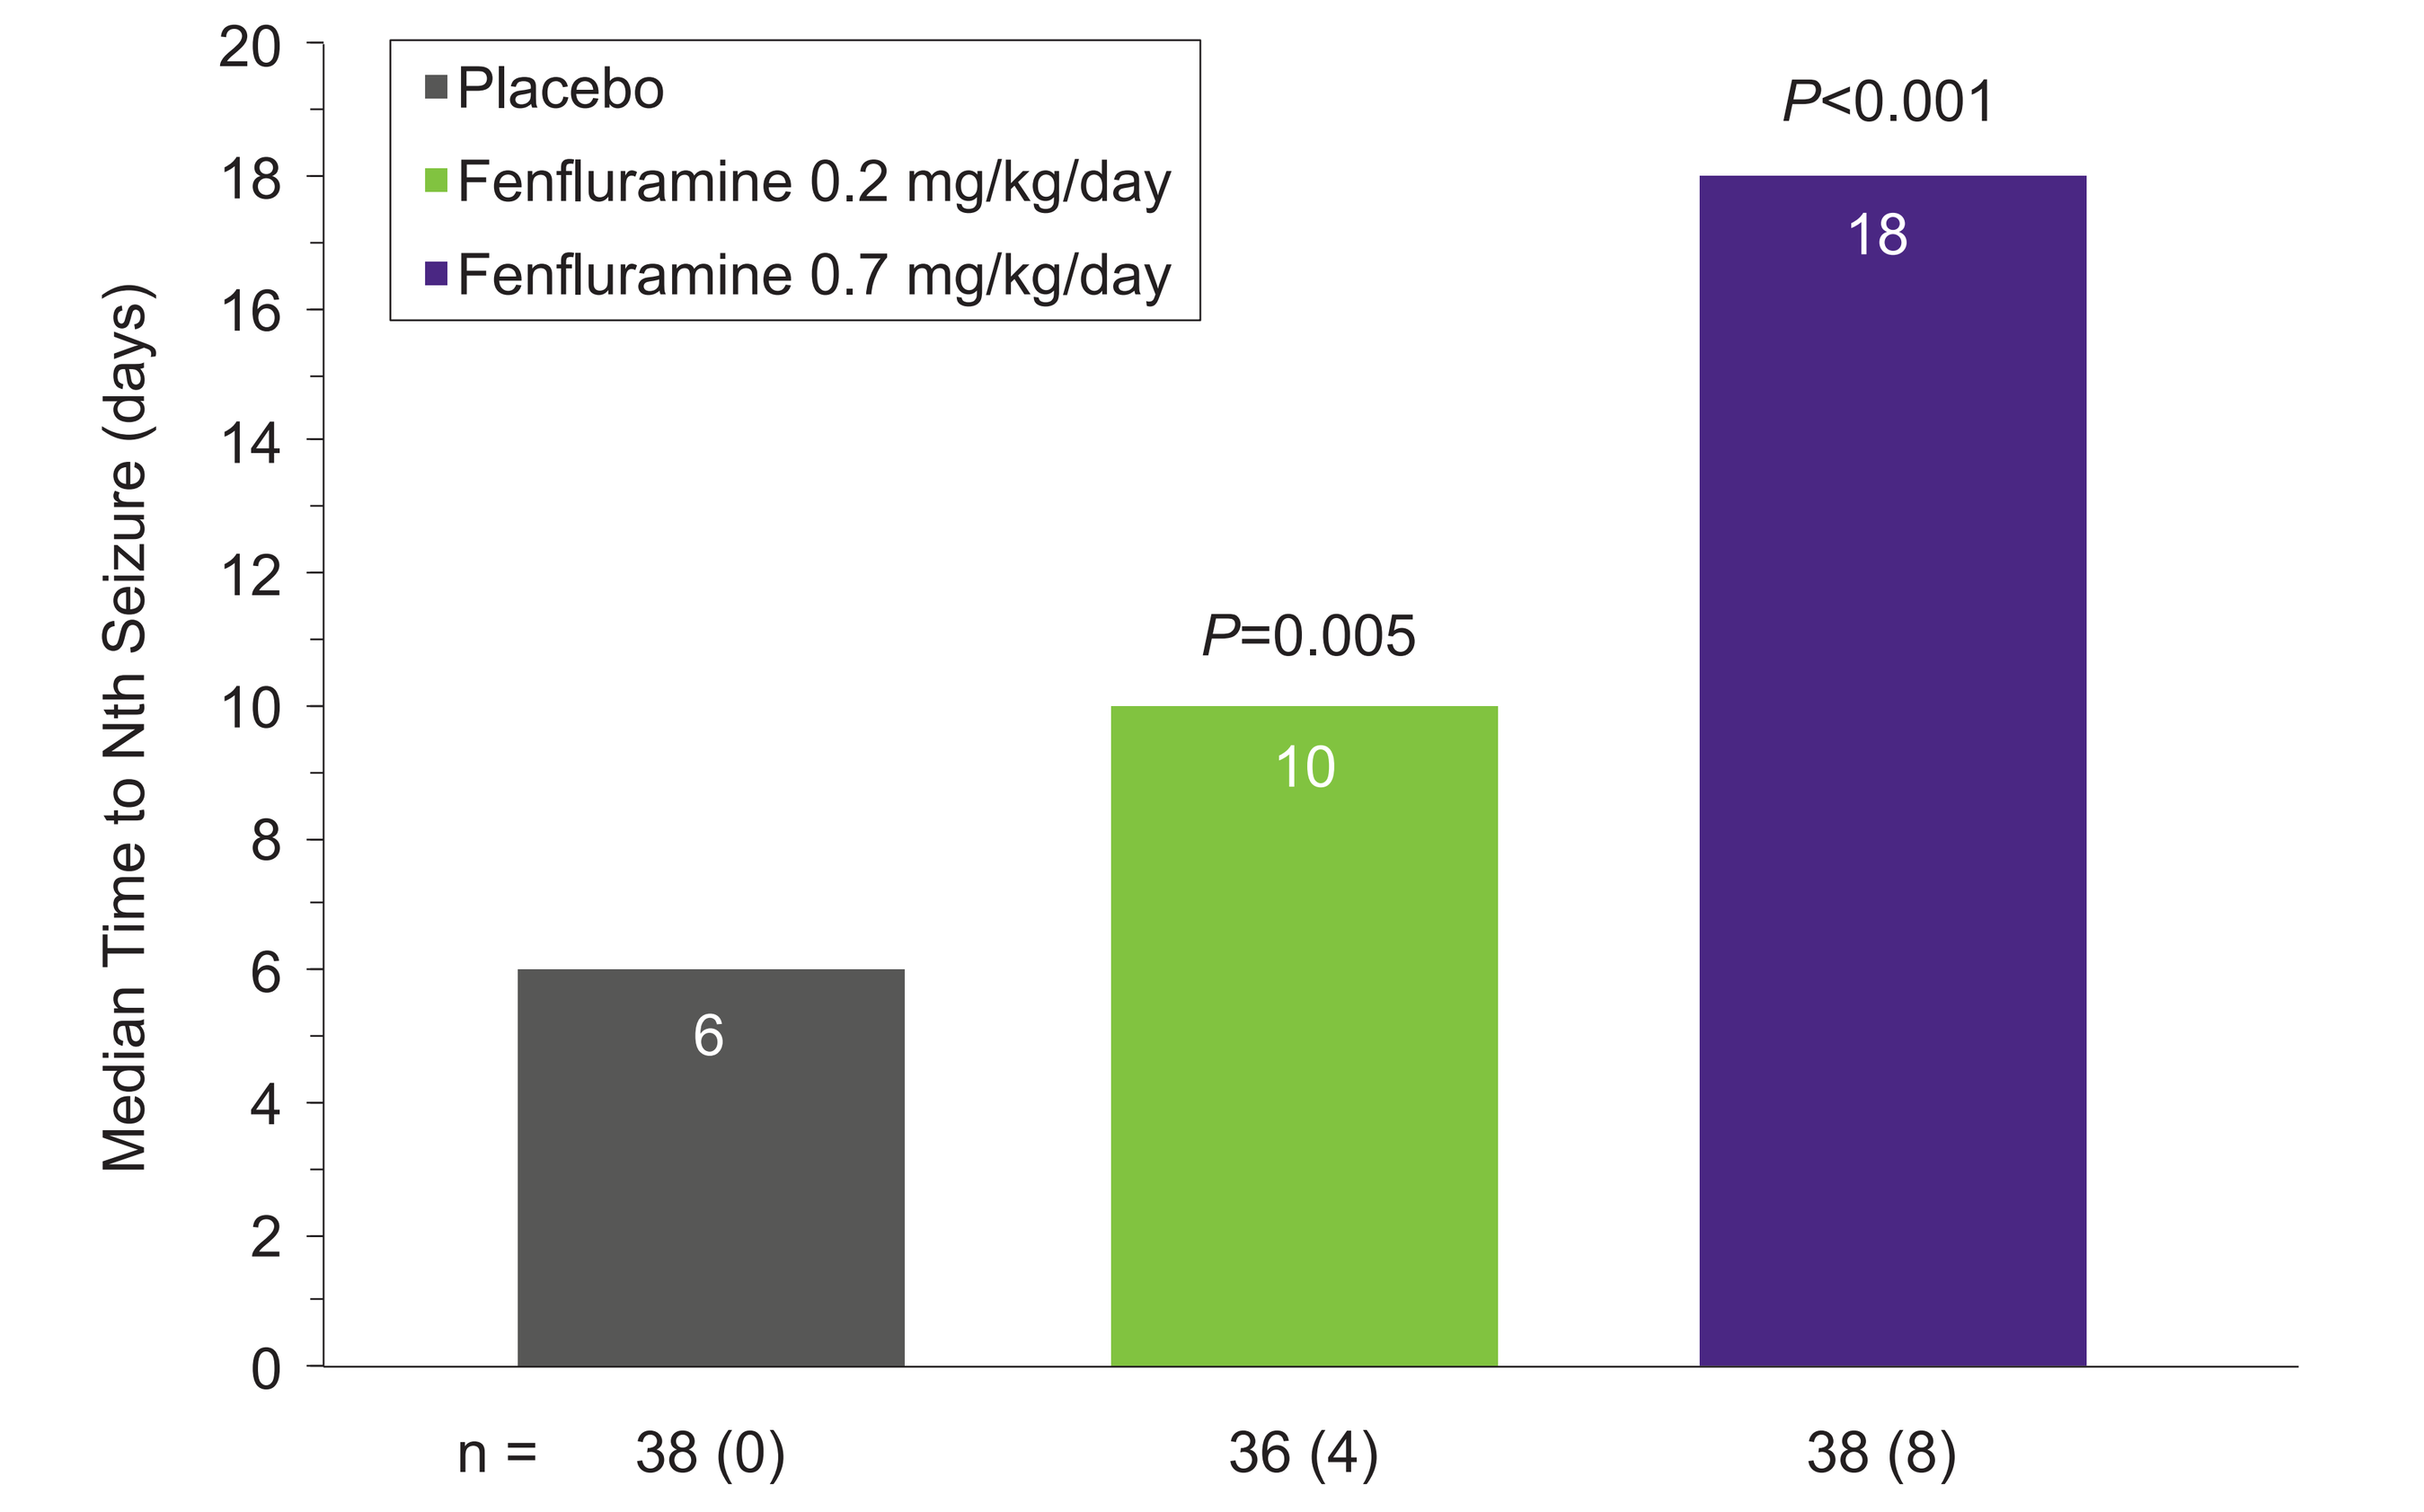

Median Time to Nth Seizure (days)
20
18
16
14
12
10
8
6
4
2
0
Placebo
Fenfluramine 0.2 mg/kg/day
Fenfluramine 0.7 mg/kg/day
6
P=0.005
10
P<0.001
18
n =
38 (0)
36 (4)
38 (8)

The number of analyzed patients in each treatment group is shown below each bar. The number in parentheses is the number of patients censored for each treatment group.

P value is for comparison with placebo.

Figure 3. Variable Baseline Period: Kaplan-Meier Curve for Time to Nth Seizure

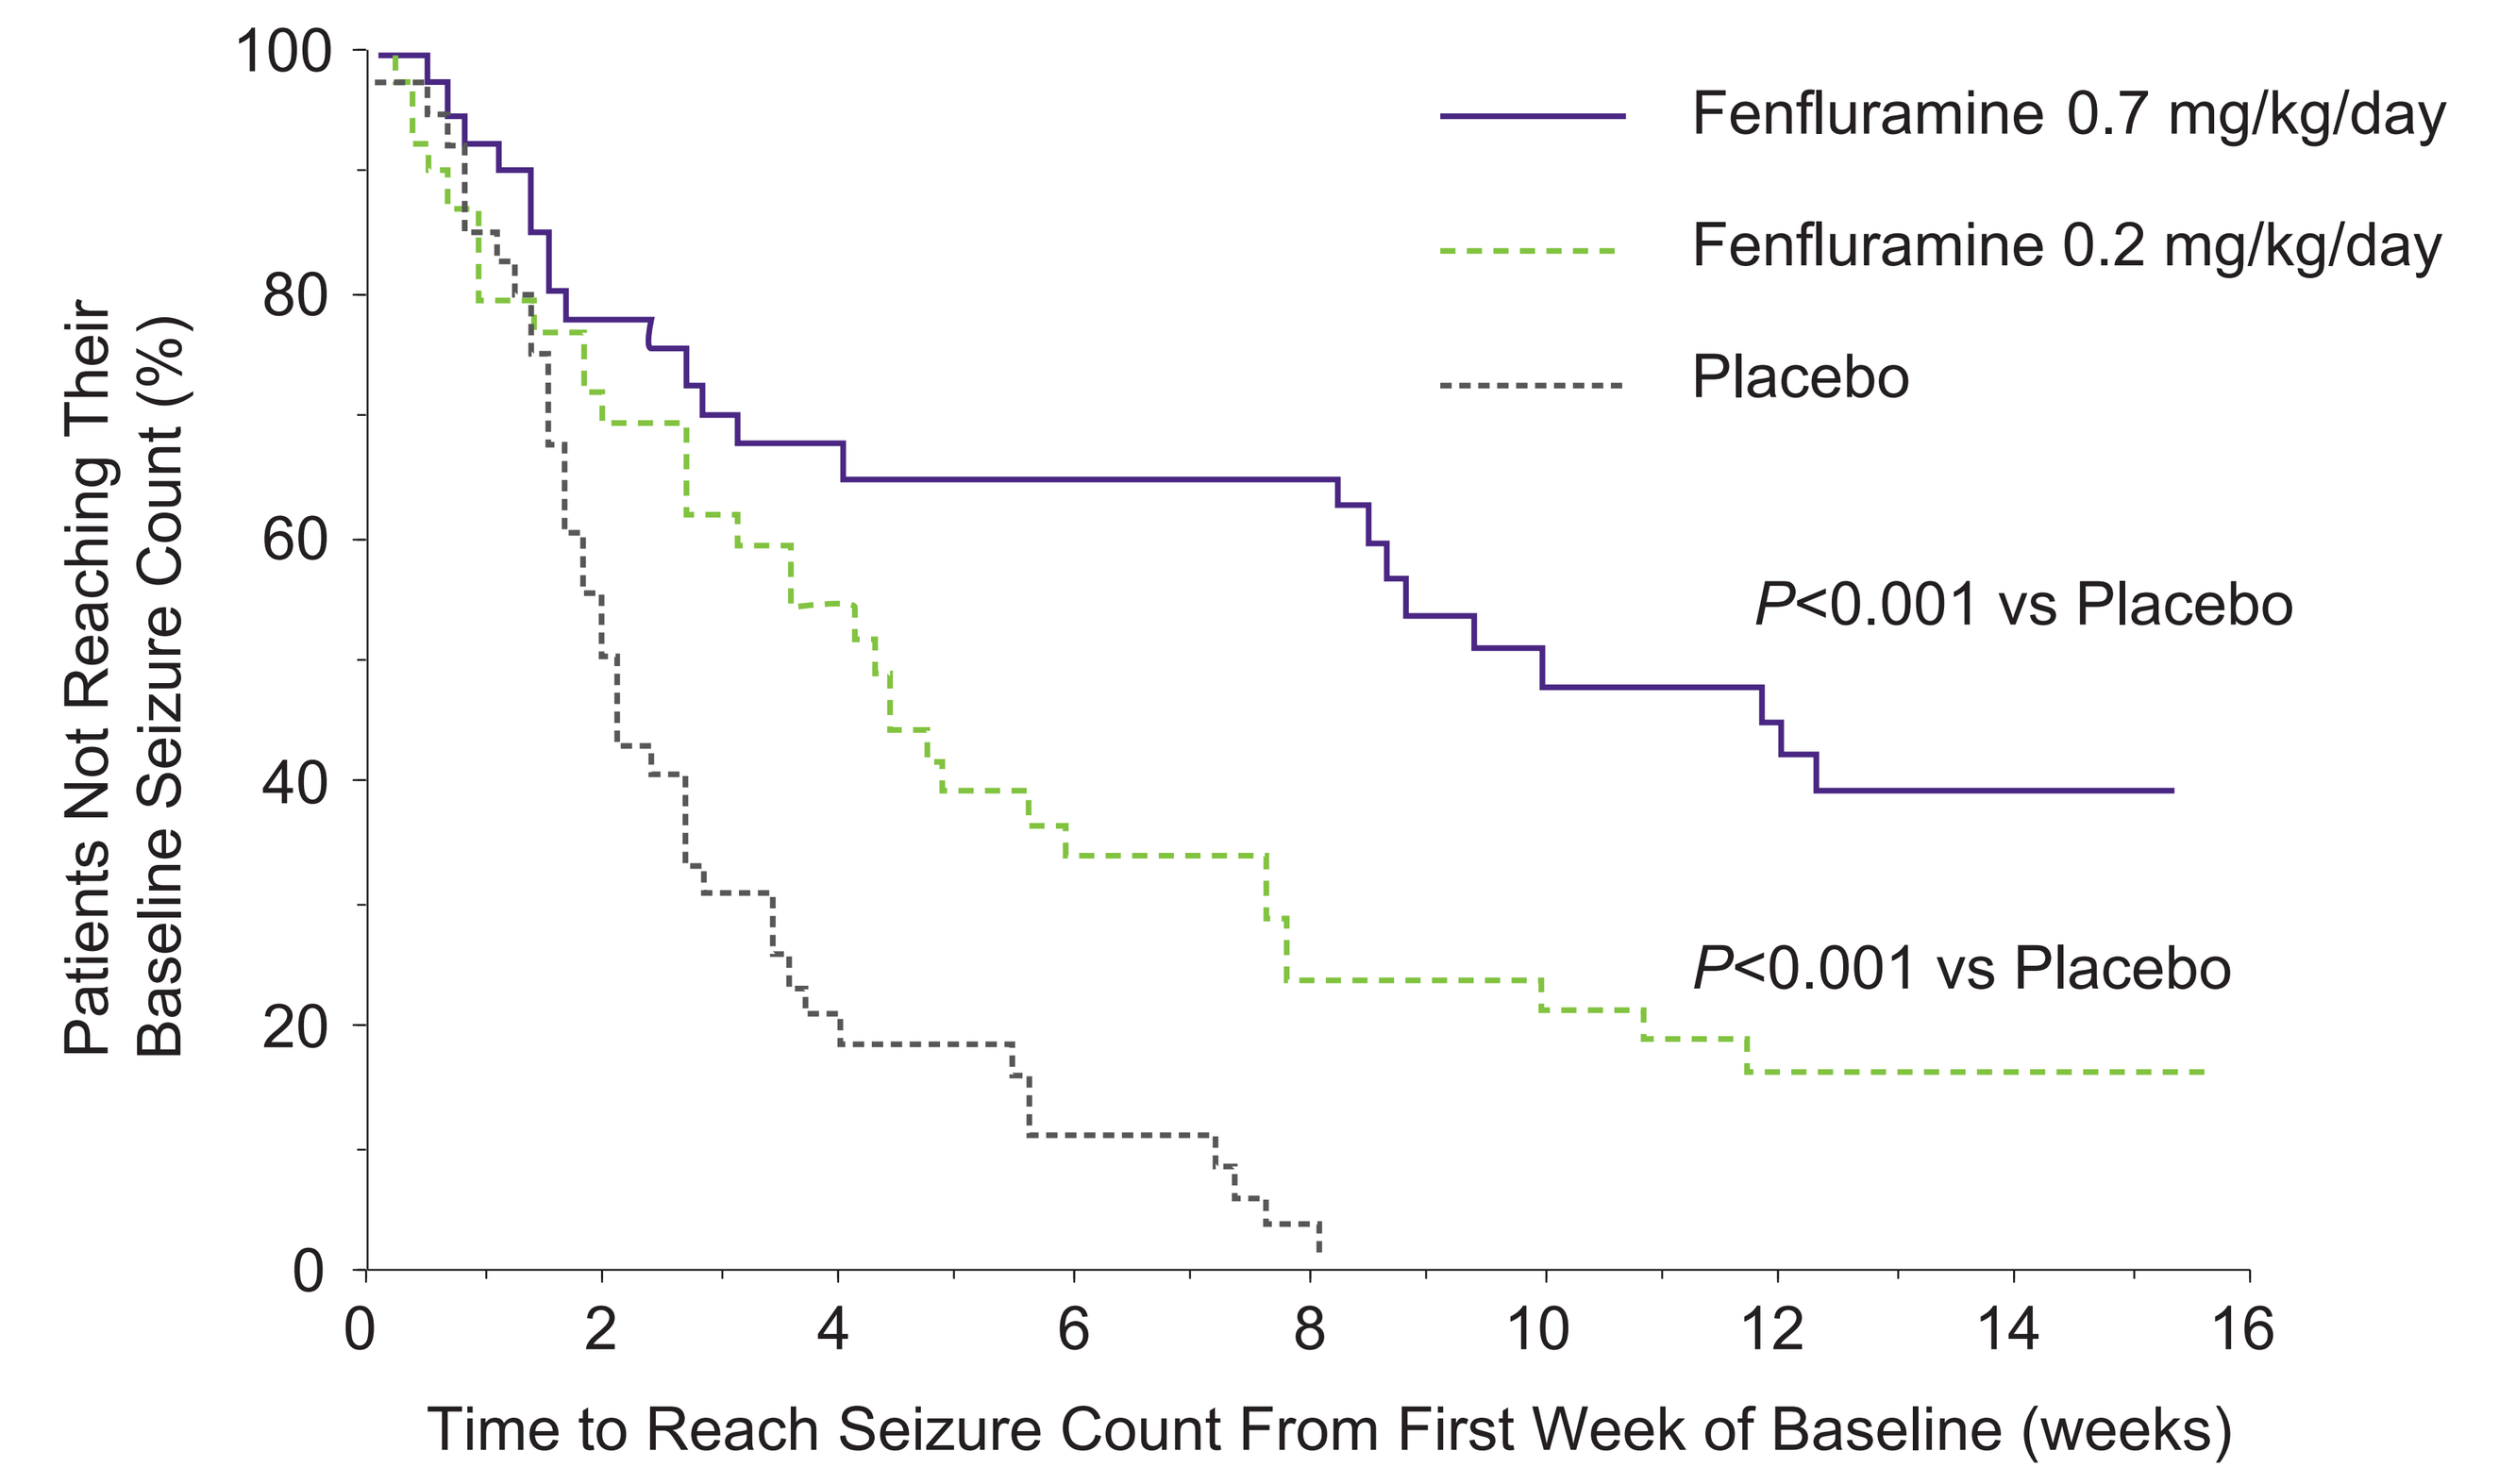

Patients Not Reaching Their Baseline Seizure Count (%)
100
80
60
40
20
0
0
2
4
6
8
10
12
14
16
Time to Reach Seizure Count From First Week of Baseline (weeks)
Fenfluramine 0.7 mg/kg/day
Fenfluramine 0.2 mg/kg/day
Placebo
P<0.001 vs Placebo
P<0.001 vs Placebo

Figure 4. Time to Nth Seizure Using a Variable Baseline Period

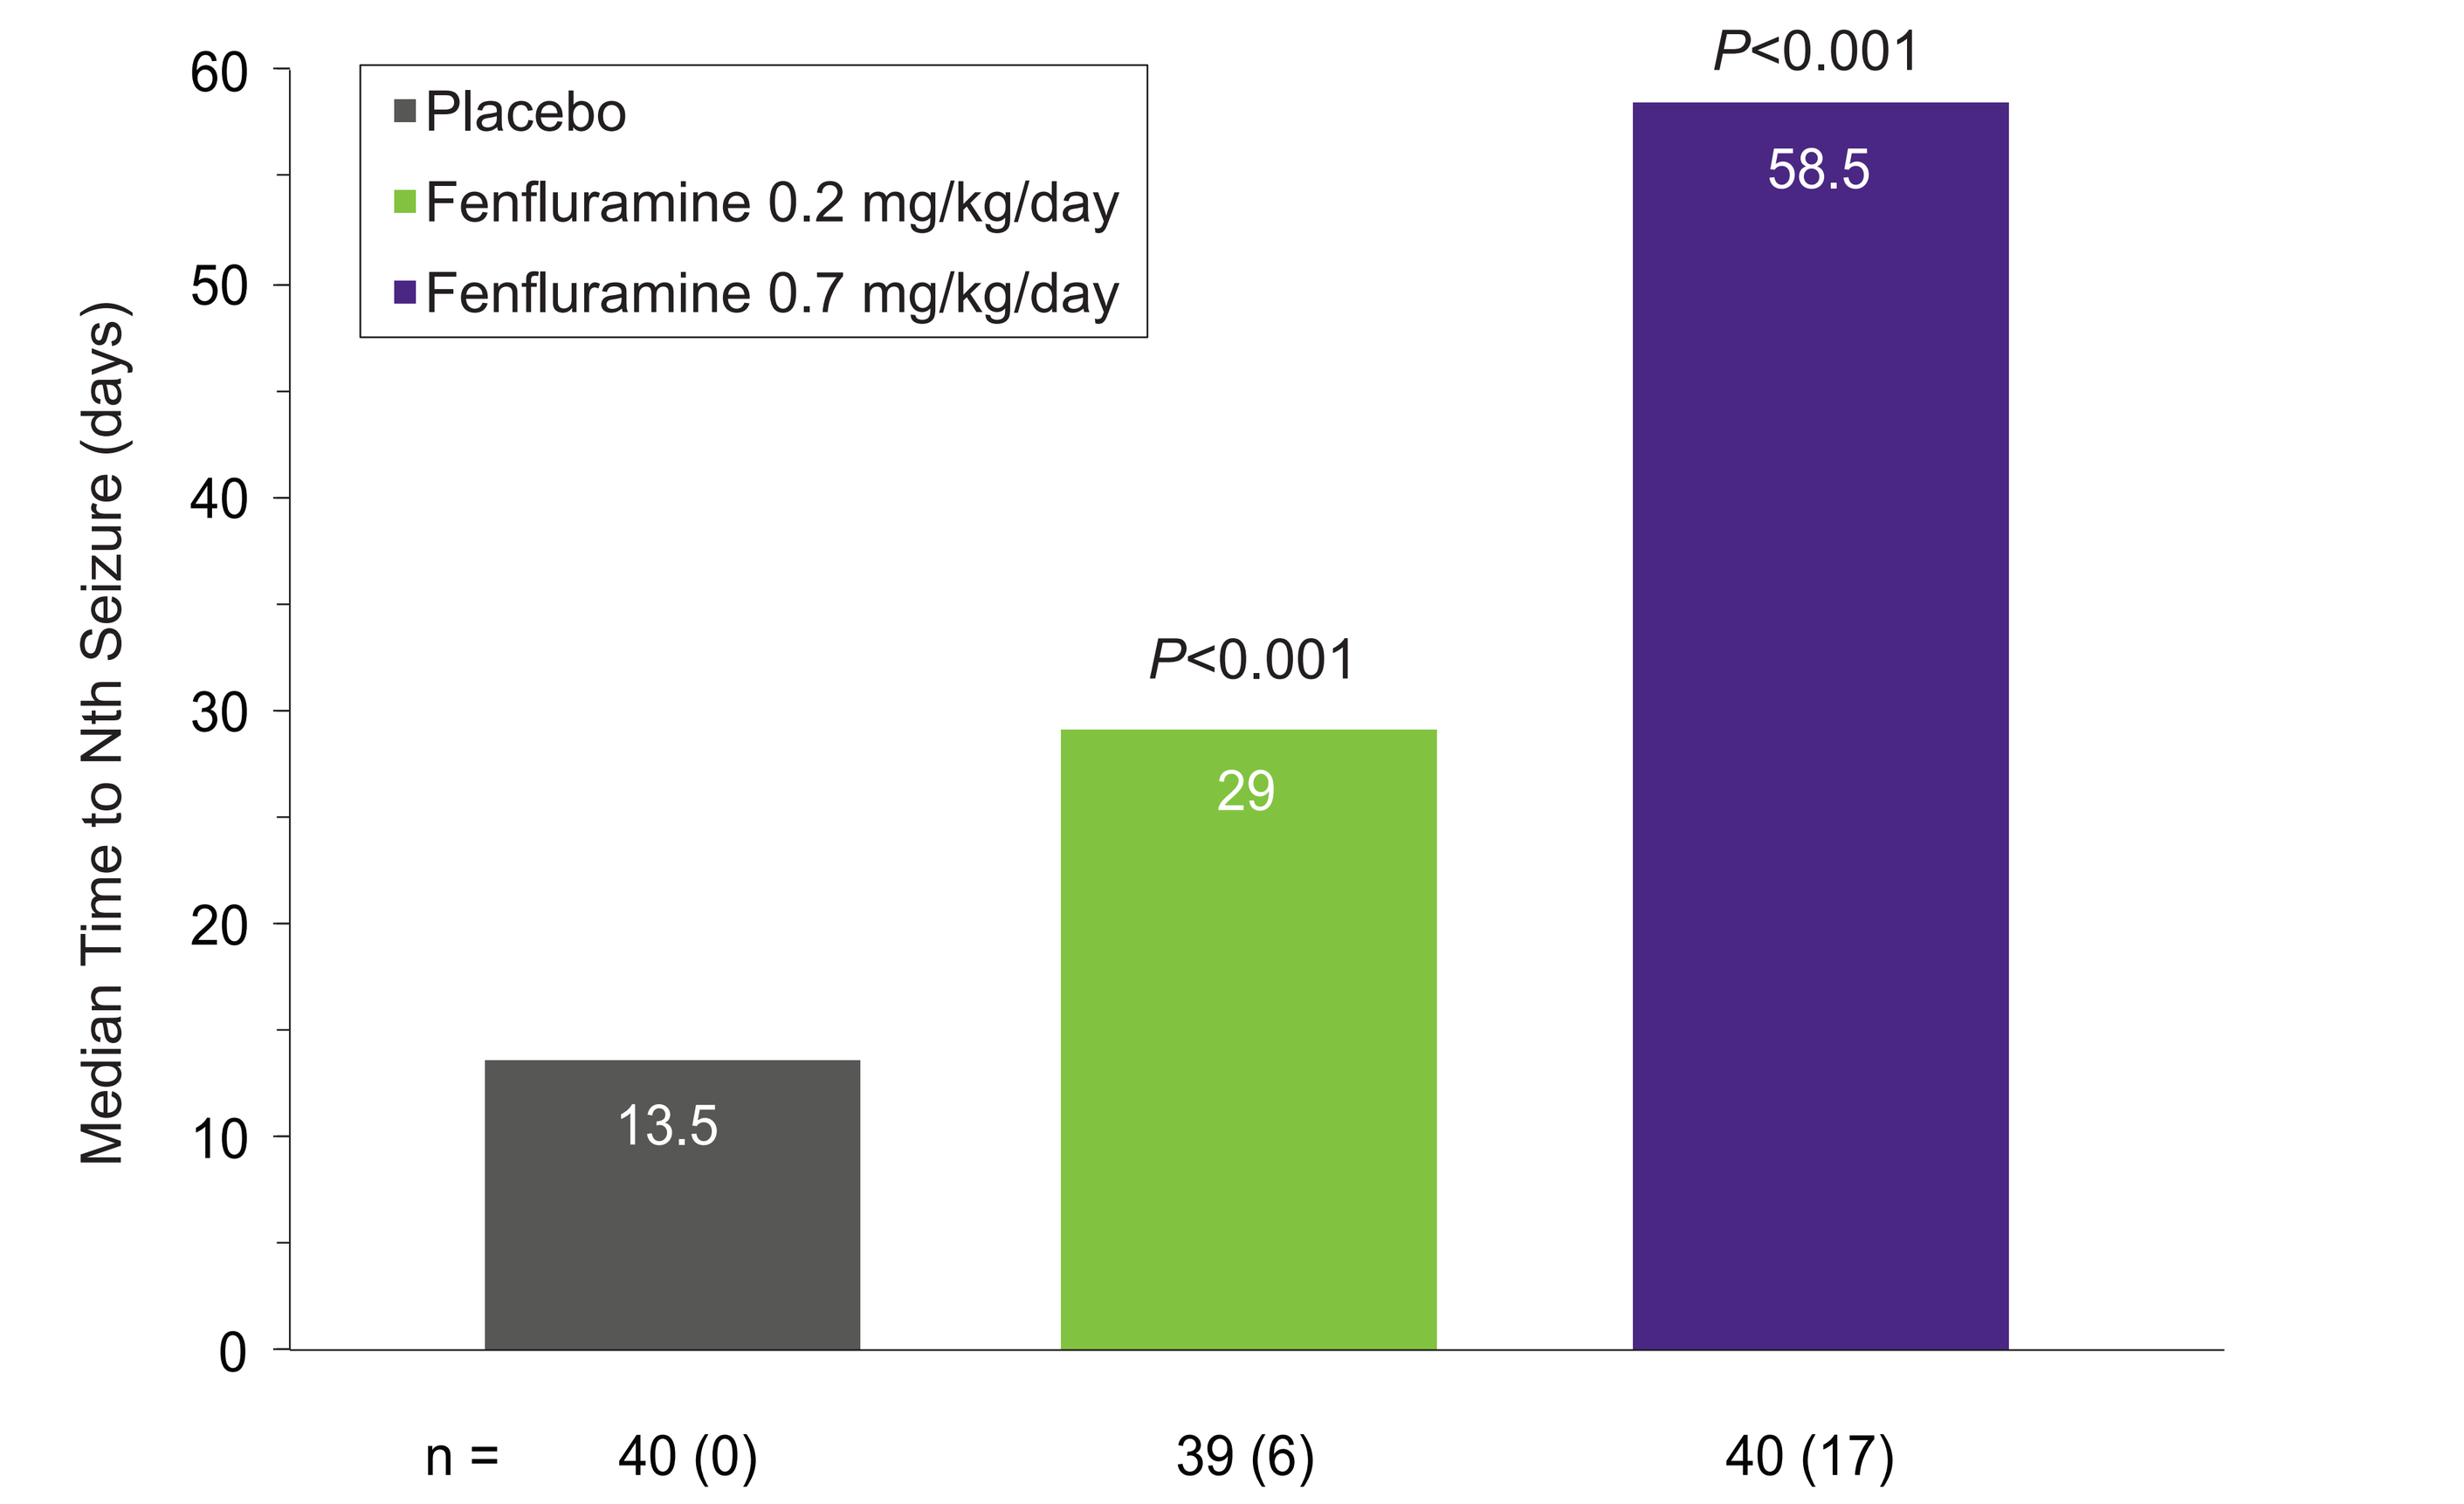

Median Time to Nth Seizure (days)
60
50
40
30
20
10
0
Placebo
Fenfluramine 0.2 mg/kg/day
Fenfluramine 0.7 mg/kg/day
13.5
P<0.001
29
P<0.001
58.5
n =
40 (0)
39 (6)
40 (17)

The number of analyzed patients in each treatment group is shown below each bar. The number in parentheses is the number of patients censored for each treatment group. The number within each bar is the median time to Nth seizure for that treatment group. The median TTE in the fenfluramine 0.7 mg/kg/day group cannot be computed exactly due to censoring; the reported median is a lower bound (ie, if it were possible to obtain more information about some of the censored observations, the median TTE in this group would be greater than or equal to the value reported here).

P value is for comparison with placebo.

Discussion

- Utilizing a 7-day baseline duration and a TTE analysis, fenfluramine demonstrated a significantly longer time to reach the Nth seizure during the treatment period: 6 days for the placebo group and 10 and 18 days for the fenfluramine 0.2 mg/kg/day (*P*=0.005), and 0.7 mg/kg/day (*P*<0.001) groups, respectively
- Similar conclusions were reached when using a variable baseline duration based on seizure frequency; 13.5, 29, and 58.5 days for the placebo, fenfluramine 0.2 mg/kg/day (*P*<0.001), and 0.7 mg/kg/day (*P*<0.001) groups, respectively
- TTE analysis also demonstrated a dose response similar to the one seen in the traditional gold standard analysis in Study 1

Conclusions

- In patient populations with a high seizure burden, a TTE analysis with a short baseline is sensitive to detect a treatment effect that is consistent with results from studies using the "gold standard" study design and analysis
- This TTE analysis with a short baseline period may reduce the duration of exposure to placebo substantially compared with the "gold standard" study design

Acknowledgments

The original phase 3 clinical study and this new analysis were sponsored by Zogenix, Inc. (Emeryville, CA, USA).

Professional medical writing and editing assistance were provided by Edward Weselcouch, PhD (PharmaWrite, LLC, Princeton, NJ, USA), and funded by Zogenix, Inc.

Disclosures

DJD: Research support, Zogenix.

JAF: Consultant and research support, Zogenix.

ML, DH: Consultant, Zogenix.

GMF, GM: Employment, Ownership interest in Zogenix, Inc.

Presented at the American Epilepsy Society (AES) Annual Meeting, December 6–10, 2019, Baltimore, MD, USA